Pharmacokinetic and Pharmacodynamic Assessment of a Lipid-based Aspirin Formulation: Results of a Prospective, Randomized, Crossover Study*

DJ. Angiolillo[1], DL. Bhatt[2], F. Lanza[3], B. Cryer[4], J. Dong[5], W. Jeske[6], E. Von chong[7], J. Prats[8], E. Deliargyris[7], U. Marathi[9]

[1]University of Florida College of Medicine – Jacksonville, Cardiology, Jacksonville, USA [2]Brigham and Women Hospital Interventional Cardiovascular Programs, Boston, USA [3]Houston Institute for Clinical Research, Houston, USA [4]University of Texas Southwestern Medical School, Dallas, USA [5]University of Washington, Hematology Division, Medicine, Seattle, USA [6]Loyola University, Cardiovascular Research Institute, Chicago Health Sciences Division, Chicago, USA [7]PLx Pharma, Inc., Houston, USA [8]Elysis, LLC., Carlisle, USA [9]7 Hills Pharma, Houston, USA

ESC Congress

Paris 2019

OBJECTIVES

Aspirin (acetylsalicylic acid), can lead to gastrointestinal mucosal injury through disruption of its protective phospholipid bilayer, underscoring the need for aspirin formulations with a more favorable safety profile while maintaining an effective pharmacologic profile.

A liquid formulation using a novel pharmaceutical lipid aspirin complex (PL-ASA) has been designed to prevent the disruption of the protective mucosal bilayer without delaying absorption.

The primary objectives of this study were to assess, for PL-ASA and traditional immediate-release aspirin (IR-ASA) at 325 mg and 650 mg doses PK and PD bioequivalence, and safety, over a 24–hour period after administration of both drugs.

METHODS

Study Design:

This was a randomized, active control, crossover study to assess bioequivalence and safety of PL-ASA vs IR-ASA administered orally. A total of 32 healthy subjects were randomized 325 or 650 mg doses of either PL-ASA or IR-ASA. After the first treatment and a minimum of a 2-week washout period, subjects were crossed over and received the alternative compound at the same dose level. Blood samples for evaluation of PK and PD were collected over a 24-hour period after each study drug administration (Figure 1).

Figure 1: Study Design and Study Population

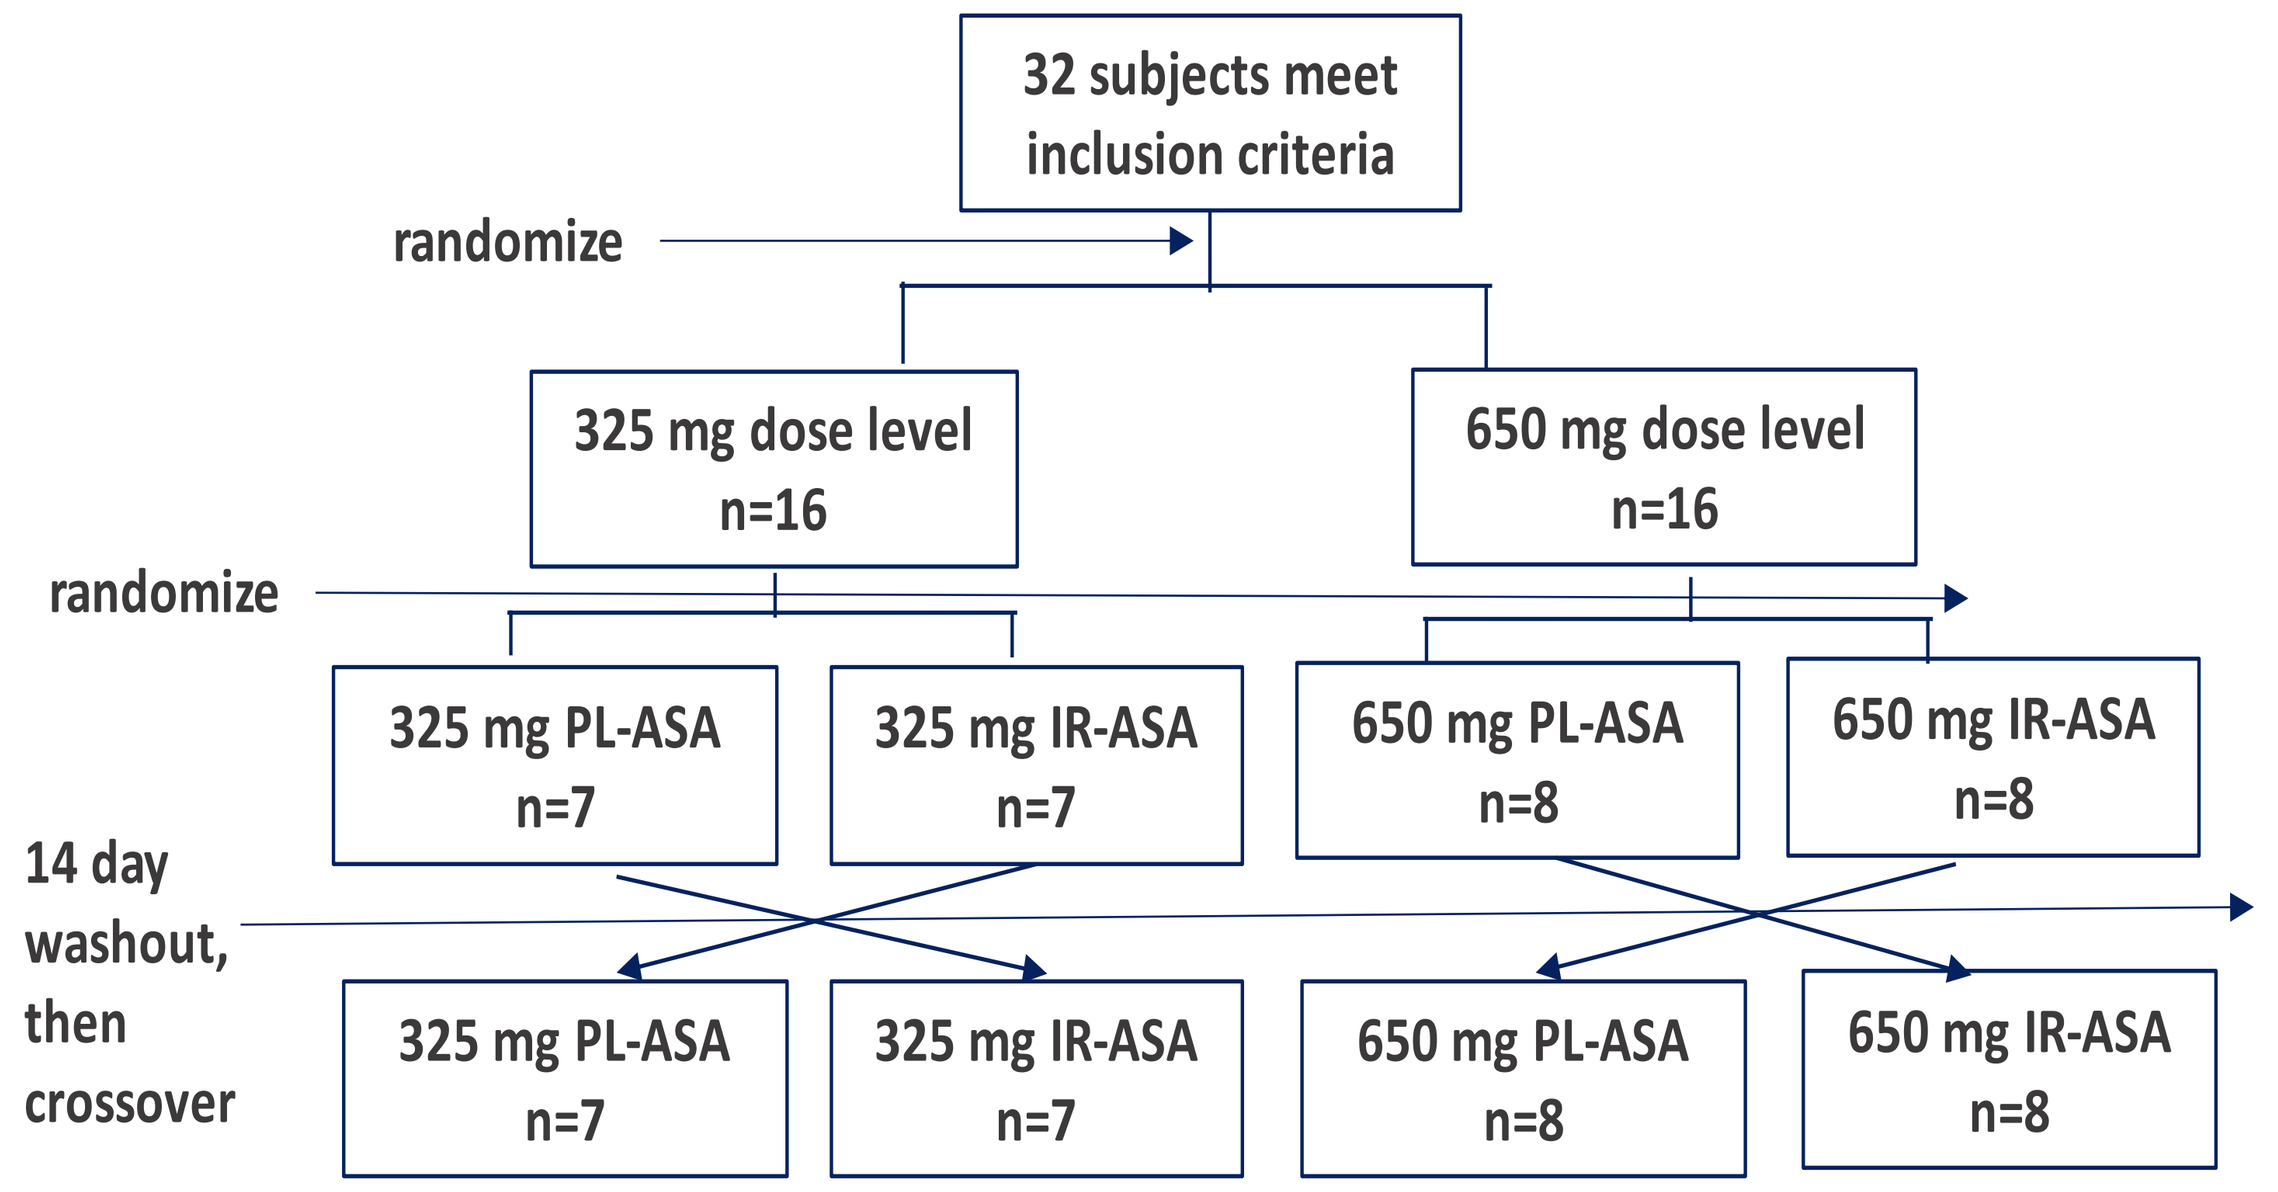

32 subjects meet inclusion criteria
randomize
325 mg dose level n=16
650 mg dose level n=16
randomize
325 mg PL-ASA n=7
325 mg IR-ASA n=7
650 mg PL-ASA n=8
650 mg IR-ASA n=8
14 day washout, then crossover
325 mg PL-ASA n=7
325 mg IR-ASA n=7
650 mg PL-ASA n=8
650 mg IR-ASA n=8

PK and PD primary endpoints:

- area-under-the-curve ($AUC_{0\text{-}t}$ and $AUC_{0\text{-infinity}}$), maximum plasma concentration (C_{max}), time to peak plasma concentration (t_{max}), terminal elimination rate constant (λ_z), first-order elimination half-life ($t_{½}$), apparent volume of distribution (V_D/F), and oral clearance (CL/F) of salicylic acid (SA),
- $AUC_{0\text{-}24}$, maximum inhibition (I_{max}), and t_{max} of % inhibition of serum thromboxane B2 (TxB2) levels

PK and PD secondary endpoints:

- $AUC_{0\text{-}t}$, $AUC_{0\text{-infinity}}$, C_{max}, t_{max}, λ_z, $t_{½}$, V_D/F, and CL/F of acetylsalicylic acid,
- incidence of aspirin responders i.e., ≥95% inhibition of serum TxB2 and urinary 11-dehydro-TxB2 ≤1500 pg/mg of creatinine,
- platelet aggregation in response to arachidonic acid (AA) and collagen

RESULTS

- A total of 32 subjects were randomized. All but two subjects, both in the 325 mg dose group, were crossed over to the second study drug (Table 1).

Table 1: Demographics of Study Population

	325-mg (n = 16)	650-mg (n = 16)	Overall (n = 32)
Age, mean ± SD	36.7 ± 9.9	36.9 ± 9.5	36.8 ± 9.6
Sex			
Male, n (%)	6 (37.5)	6 (37.5)	12 (37.5)
Female, n (%)	10 (62.5)	10 (62.5)	20 (62.5)
Ethnicity			
White, n (%)	8 (50.0)	10 (62.5)	18 (56.3)
Native Hawaiian/Islander, n (%)	1 (6.2)	0 (0)	1 (3.1)
African American, n (%)	7 (43.8)	6 (37.5)	13 (40.6)
Height (inches)	66.5 ± 4.3	68.8 ± 3.7	67.7 ± 4.1
Weight (pounds)	167.4 ± 55.7	174.3 ± 39.2	170.8 ± 47.5
Subjects Completed Study Per Protocol	14 (87.5%)	16 (100%)	30 (93.8%)
Unable to confirm baseline platelet aggregation after washout	2 (12.5%)	0 (0%)	2 (6.2%)

- The salicylic acid concentration-time curves for both PL-ASA and IR-ASA at both dose levels were very similar, but the peak concentration was nominally slightly higher and elimination from blood was faster after PL-ASA (Table 2, Figure 2).

Table 2: Salicylic Acid PK Parameters after Drug Administration

325-mg dose*	PL-ASA (n=13)		IR-ASA (n=13)	
Parameter	median	range	median	range
$AUC_{0\text{-}t}$ (μg×min/mL)	5489	2968 – 10795	5401	2736 –13855
$AUC_{0\text{-}\infty}$ (μg×min/mL)	5501	3123 – 13239	5801	2863 –13964
C_{max} (μg /mL)	19	10 – 27	16	1 – 25
t_{max} (min)	120	75 – 240	120	75 – 240
λ(1/min)	0.005	0.003 – 0.007	0.005	0.002 –0.007
$t_{½}$ (min)	143	98 – 249	151	99 – 353

650-mg dose†	PL-ASA (n=14)		IR-ASA (n=14)	
Parameter	median	range	median	range
$AUC_{0\text{-}t}$ (μg×min/mL)	14834	8248 – 23332	15444	7673 – 22429
$AUC_{0\text{-}\infty}$ (μg ×min/mL)	14856	8847 – 23515	15477	8150 – 22926
C_{max} (μg /mL)	35	25 – 53	36	24 – 44
t_{max} (min)	180	120 – 360	180	75 – 240
λ_z(1/min)	0.005	0.003 – 0.006	0.005	0.003 – 0.006
$t_{½}$ (min)	136	117 – 250	150	119 – 270

* PK population does not include 2 subjects who did not receive planned treatment, and 1 subject whose dosing was not as protocol-specified. †two subjects were excluded whose dosing was not as protocol-specified.
$AUC_{0\text{-}t}$ =area-under-the-curve, $AUC_{0\text{-}\infty}$ = $AUC_{0\text{-}t}$ extrapolated to infinity, C_{max} =maximum plasma concentration, μg=micrograms, min=minutes, mL=milliliters, n=number, t_{max} =time of peak drug concentration, λ_z =terminal elimination rate constant, $t_{½}$ =first-order elimination half-life

Figure 2: Impact of PL-ASA and IR-ASA Formulations on Aspirin's Disposition

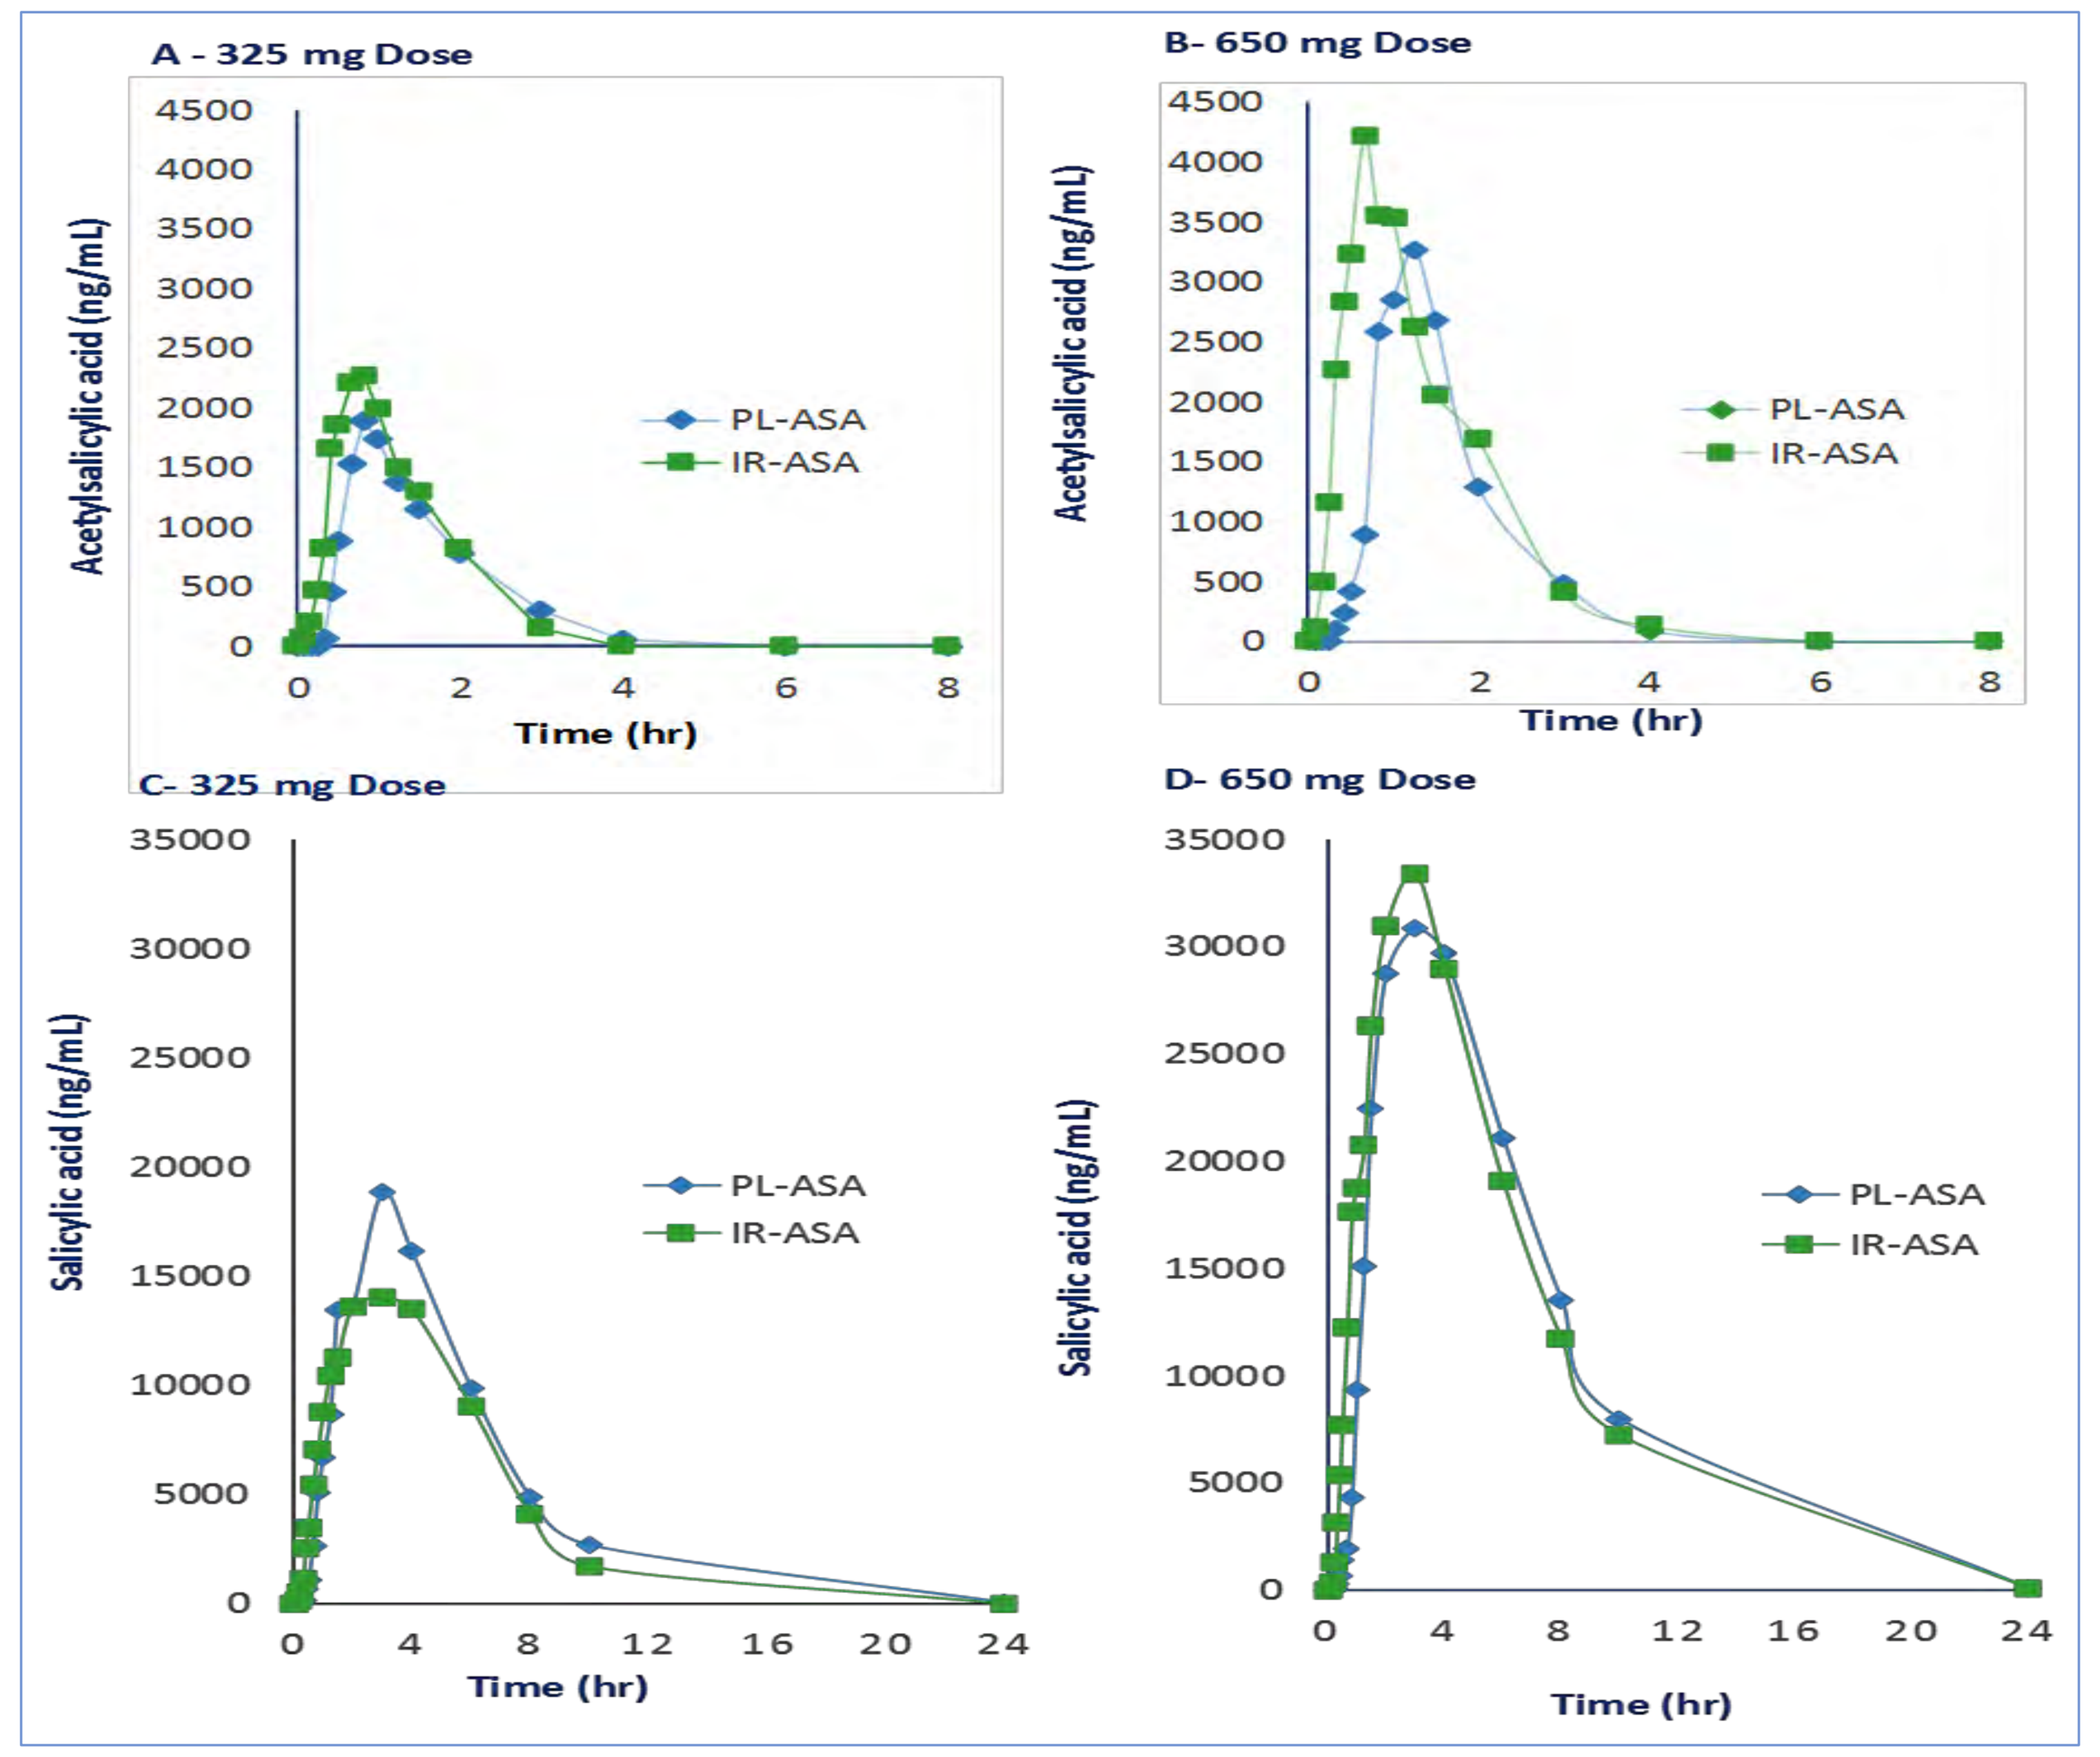

A - 325 mg Dose
B- 650 mg Dose
C- 325 mg Dose
D- 650 mg Dose
Acetylsalicylic acid (ng/mL)
Salicylic acid (ng/mL)
Time (hr)
PL-ASA
IR-ASA

- PK and PD equivalence of PL-ASA to IR-ASA was based on log-transformed ratios for $AUC_{0\text{-}t}$, $AUC_{0\text{-}\infty}$, and C_{max} of plasma salicylic acid and for $AUC_{0\text{-}24}$ and I_{max} of the percent inhibition of serum TxB2 levels.
- Bioequivalence analyses showed that 90% CIs for geometric mean of salicylic acid parameters $AUC_{0\text{-}t}$, $AUC_{0\text{-}\infty}$, and C_{max} for the ratio PL-ASA and IR-ASA at both dose levels were within 80% and 125%, indicating the nearly the same amount of SA was metabolized by each subject (Table 3).

Table 3: PK Bioequivalence of PL-ASA and IR-ASA

Salicylic acid 325-mg dose (n=13)	Ratio (%)†	90% CI‡	P - value§
$AUC_{0\text{-}t}$ (μg×min/mL)	97	89 – 104	0.43
$AUC_{0\text{-infinity}}$ (μg ×min/mL)	98	91 – 106	0.62
C_{max} (μg /mL)	104	92 – 117	0.59
Salicylic acid 650-mg dose (n=14)			
$AUC_{0\text{-}t}$ (μg×min/mL)	98	93 – 103	0.44
$AUC_{0\text{-infinity}}$ (μg ×min/mL)	99	95 – 103	0.65
C_{max} (μg /mL)	106	97 – 115	0.25

* Only subjects who received both treatments and whose appropriate dosing was verified are included
† Ratio = 100 × Geometric Mean (PL-ASA) / Geometric Mean (IR- ASA)
‡ 90% Confidence Interval on the Ratio of PL-ASA to IR-ASA § ANOVA p-value for the difference in the treatment estimates.

- The mean concentration of serum TxB2 over time following dosing with PL-ASA and IR-ASA are similar (Figure 3). In addition, C_{min} (TxB2) values for both drugs were below 3.1 ng/mL, suggesting that PL-ASA and IR-ASA could be considered functionally and clinically equivalent. However, the time required to reach this 3.1 ng/mL limit was shorter following PL-ASA at the 650 mg dose level.

Figure 3: Mean Concentration of Serum TxB2

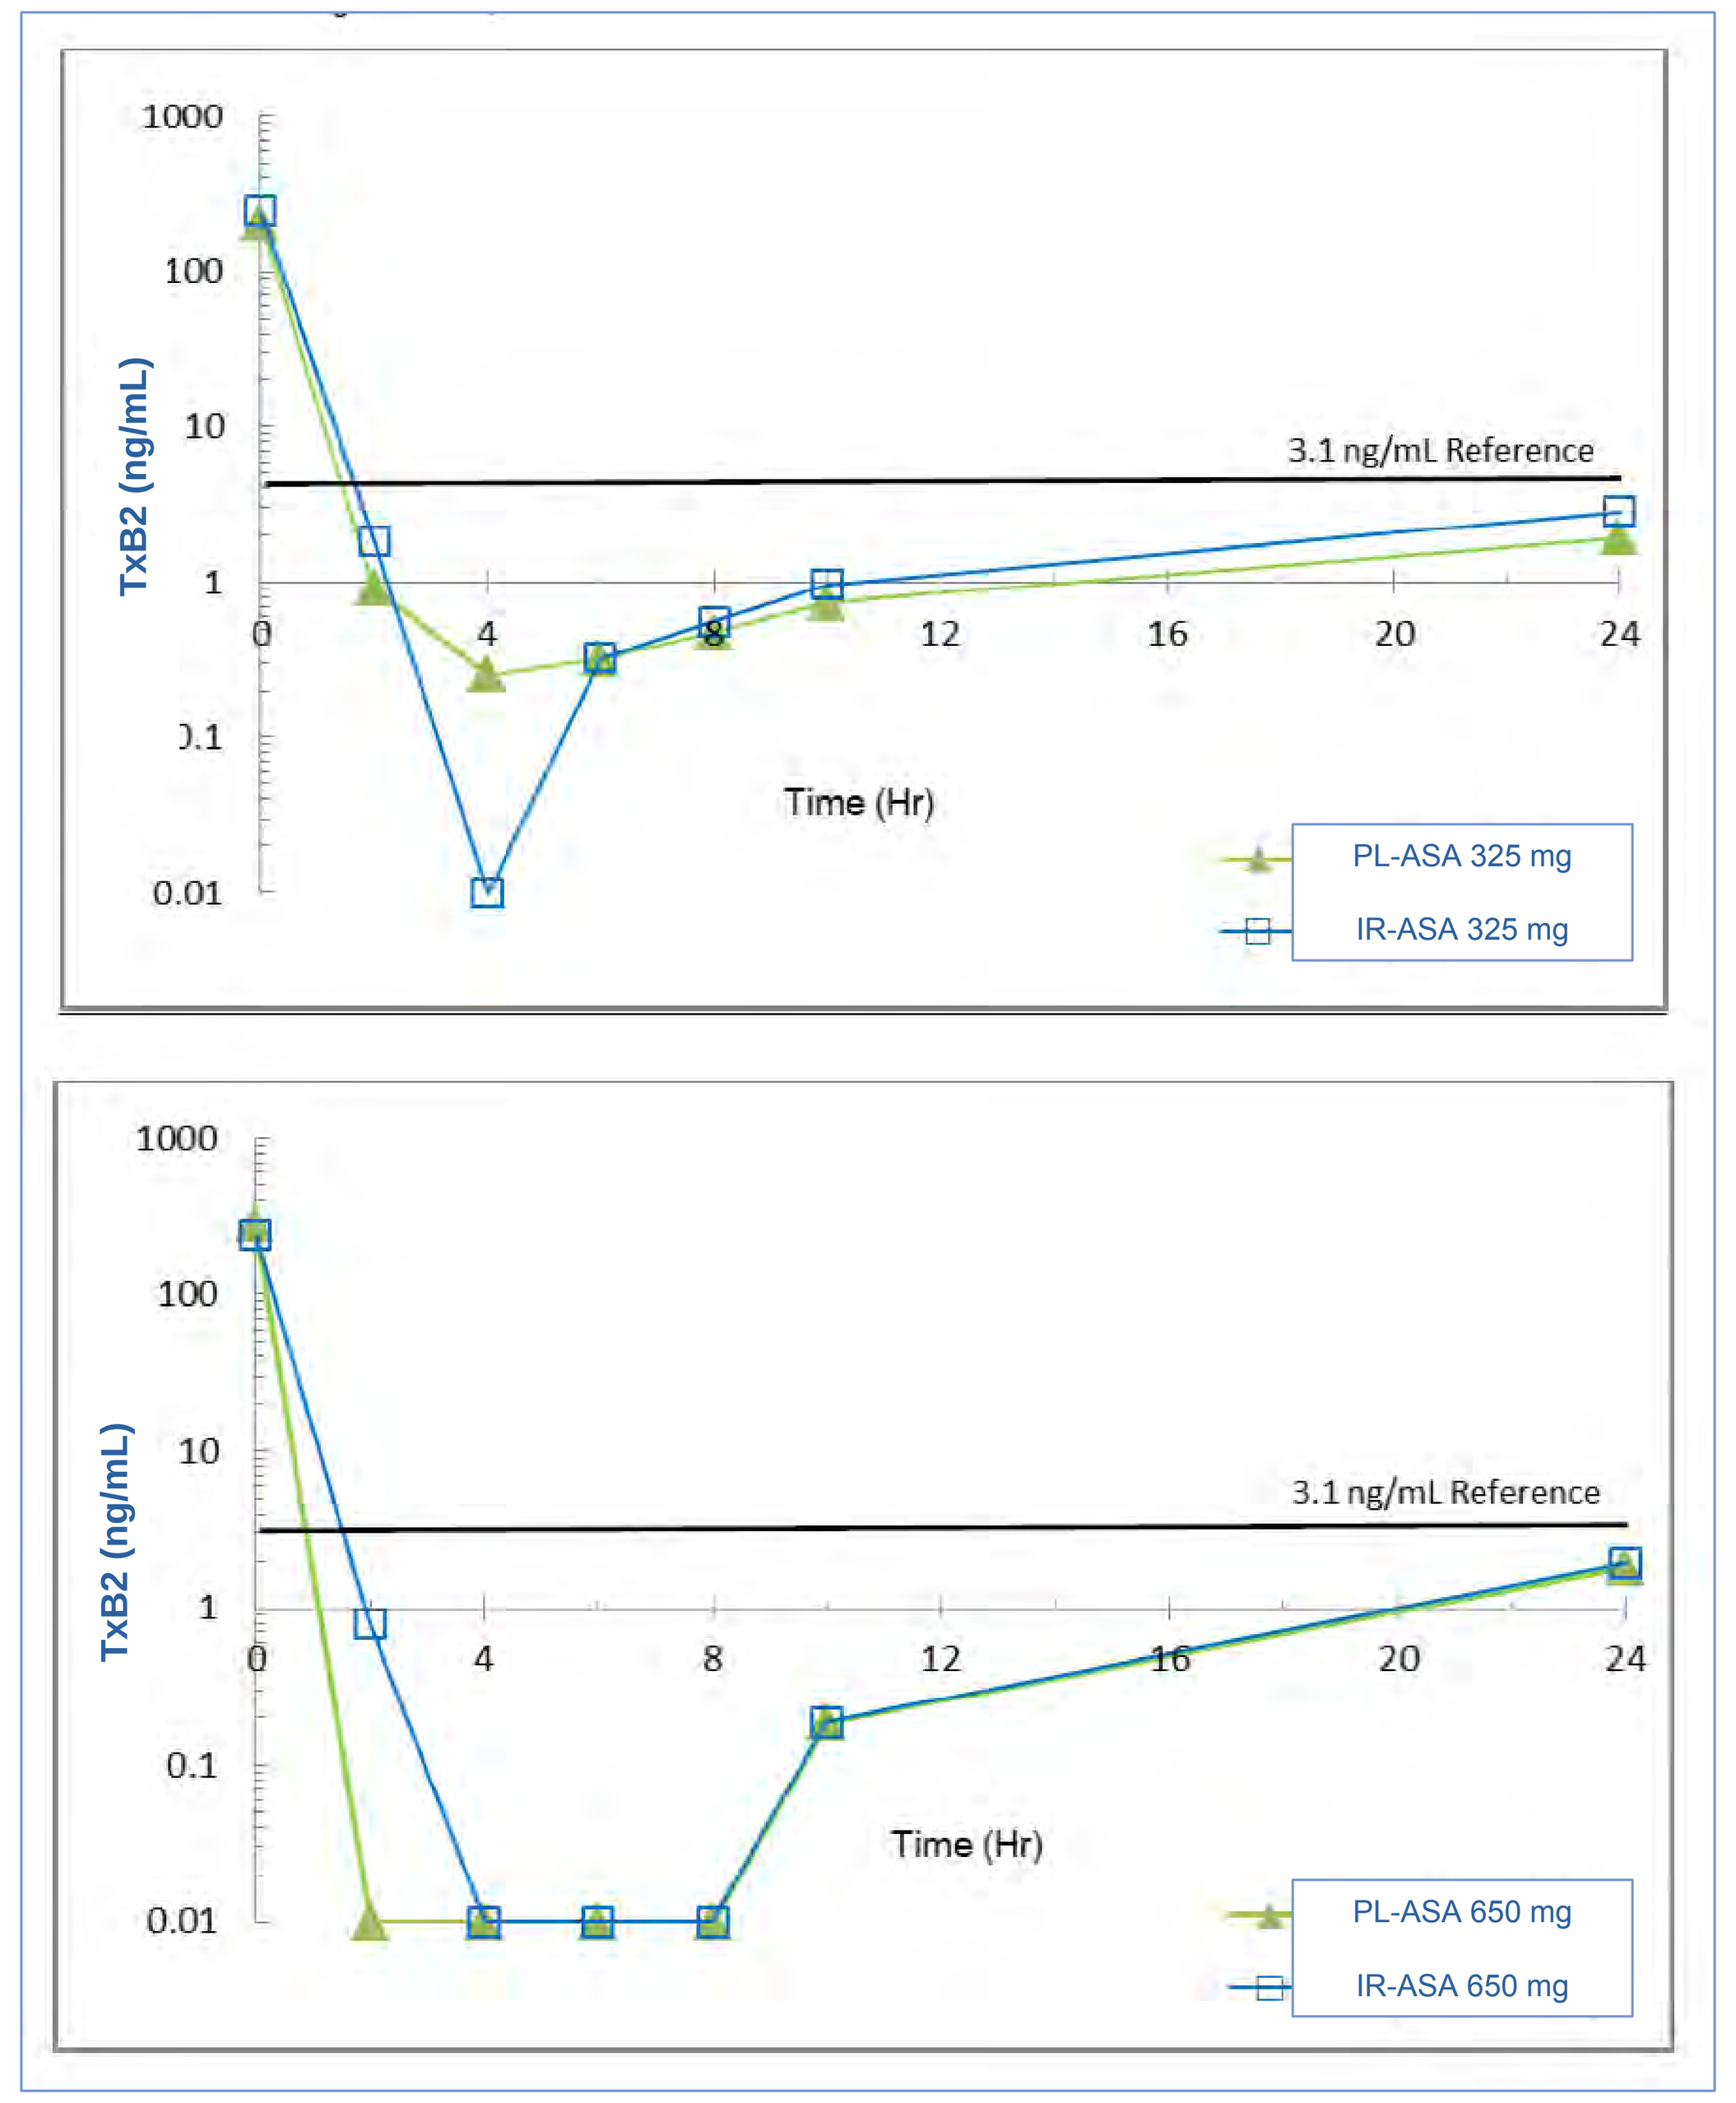

TxB2 (ng/mL)
Time (Hr)
3.1 ng/mL Reference
PL-ASA 325 mg
IR-ASA 325 mg
PL-ASA 650 mg
IR-ASA 650 mg

- The 90% CIs at both dose levels for the mean log-transformed parameters of % inhibition of TxB2 were within 80% and 125% bioequivalence interval (Data not shown).
- Platelet aggregation in response to arachidonic acid in platelet-rich plasma from subjects at baseline, and 6 and 24 hours after PL-ASA or IR-ASA treatment at either dose level was >99%. Collagen-induced aggregation was inhibited to a lesser, but similar degree for both drugs.
- All subjects were aspirin responders (≥99% inhibition of serum TxB2). Urinary 11-dehydro-TxB2 assay results showed that 85.7% of subjects were responders to IR-ASA and 78.6% were responders to PL-ASA at 325 mg doses; 100.0% responded to IR-ASA and 93.8% to PL-ASA at 650 mg doses.
- PL-ASA was safe; only 1 unrelated, mild adverse event (AE) was reported; no serious AE; and no clinically significant vital sign or lab abnormalities occurred.

CONCLUSIONS

- PK parameters were similar for PL-ASA and IR-ASA, and met FDA-criteria for bioequivalence at both 325-mg and 650-mg doses.
- 90% CIs for the mean log-transformed salicylic acid PK parameters were within the bioequivalence acceptance interval for both 325 mg and the 650 mg doses.
- All PK parameters for plasma acetylsalicylic acid levels were within the 80% to 125% bioequivalence range at the 325 mg dose level; at the 650 mg dose, the 90% CIs for the mean log-transformed acetylsalicylic acid parameters $AUC_{0\text{-}t}$ and $AUC_{0\text{-infinity}}$ were within the range.
- With respect to PD, PL-ASA and IR-ASA had similar profiles at the 325 mg and 650 mg doses. Both drugs also showed C_{min} TxB2 values <3.1 ng/mL (cut-off associated with decreased cardiovascular events) and >99% inhibition of serum TxB2 (≥95% inhibition represents the cut-off for aspirin responders).
- PD equivalence was further supported by the observation of complete inhibition of arachidonic acid-induced platelet aggregation following both PL-ASA and IR-ASA administration at both dose levels.
- Secondary PK/PD parameters showed similar results
- Administration of a single dose of PL-ASA at 325-mg or 650-mg is safe.
- The findings of this study demonstrate that PL-ASA is both bioequivalent to aspirin for non-prescription indications, and pharmacodynamically equivalent to aspirin for physician-directed cardiovascular indications.
- The improved endoscopic safety profile of PL-ASA coupled with its pharmacologic efficacy equivalent to IR-ASA may result in an improved benefit-risk profile. Further studies are warranted to test the performance of PL-ASA versus enteric-coated aspirin, the aspirin preferred by the majority of physicians and patients.

Declaration of Interest

PLx Pharma Inc. was the study sponsor. The project described was supported by Grant Number R42DK063882 from the National Institute Of Diabetes And Digestive And Kidney Diseases. The content is solely the responsibility of the authors and does not necessarily represent the official views of the NIDDKD or the NIH.

Drs. Angiolillo, Bhatt, and Cryer have received consulting fees from PLx Pharma Inc. Dr. Prats is a consultant to PLx Pharma Inc. Dr Marathi, Dr. Deliargyris and Ms. Von Chong are employees of PLx Pharma Inc. All other authors had no conflicts related to the current study.

Corresponding Author

Dominick J. Angiolillo, MD. E-mail: dominick.angiolillo@jax.ufl.edu

***ClinicaTrials.gov: NCT04008979**